63rd ERA CONGRESS
GLASGOW & VIRTUAL
JUNE 3-6, 2026
Challenge Your Thinking

A Physiology-Guided Strategy to Address Metabolic Acidosis in Chronic Kidney Disease: Rationale and Design of the REVIVE Study

B. SINGH [1], **G. CHERTOW** [2], **W. RENDER** [1], **S TURNER** [1], **and R. GLASSOCK** [3]

1 Renibus Therapeutics, Southlake, Texas, USA, 2 Stanford University, Palo Alto, California, USA, 3 Geffen School of Medicine at UCLA, Los Angeles, California, USA

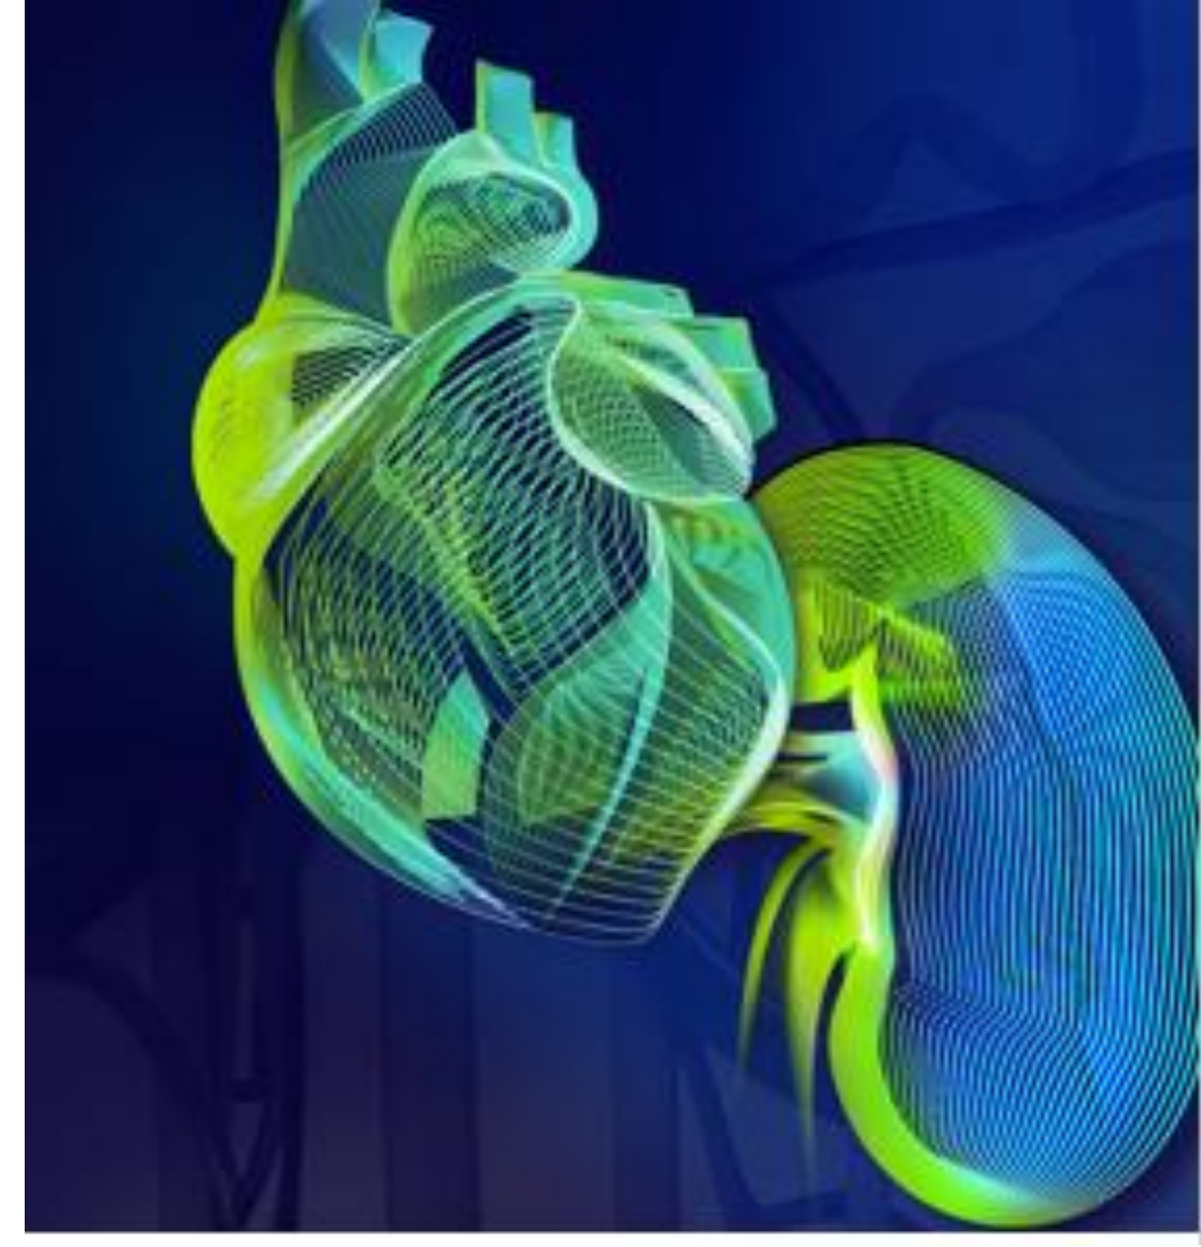

INTRODUCTION

- Metabolic acidosis is a common complication of chronic kidney disease (CKD), affecting 15-50% of patients depending upon CKD stage
- Metabolic acidosis contributes to CKD progression, sarcopenia, osteopenia, and reduced quality of life, yet remains under-recognized and under-treated
- Current management includes alkali treatment which is limited by tolerability, sodium/potassium load, high pill burden, and poor adherence
- Veverimer is a non-absorbed polymer that binds hydrochloric acid (HCl) in the stomach, thereby increasing serum bicarbonate concentration (SBC) without counterion exposure[1]
- Prior studies with veverimer demonstrated improvements in SBC, although variability in response suggested that gastric physiologic factors may influence treatment efficacy [2,3]

AIM

- Identify gastrointestinal physiologic factors that may influence treatment efficacy of veverimer

METHODS

- We performed translational analyses incorporating prior non-clinical and clinical trial data, gastric physiology, and pharmacologic modeling to identify determinants of veverimer's efficacy
- Evaluated factors included: HCl concentrations at various gastric pH levels, gastric acid volume, gastric emptying time, meal composition, dosing frequency, and impact of medications/ medical conditions known to reduce gastric acid secretion

RESULTS

A. Veverimer MOA and Gastric HCl Binding

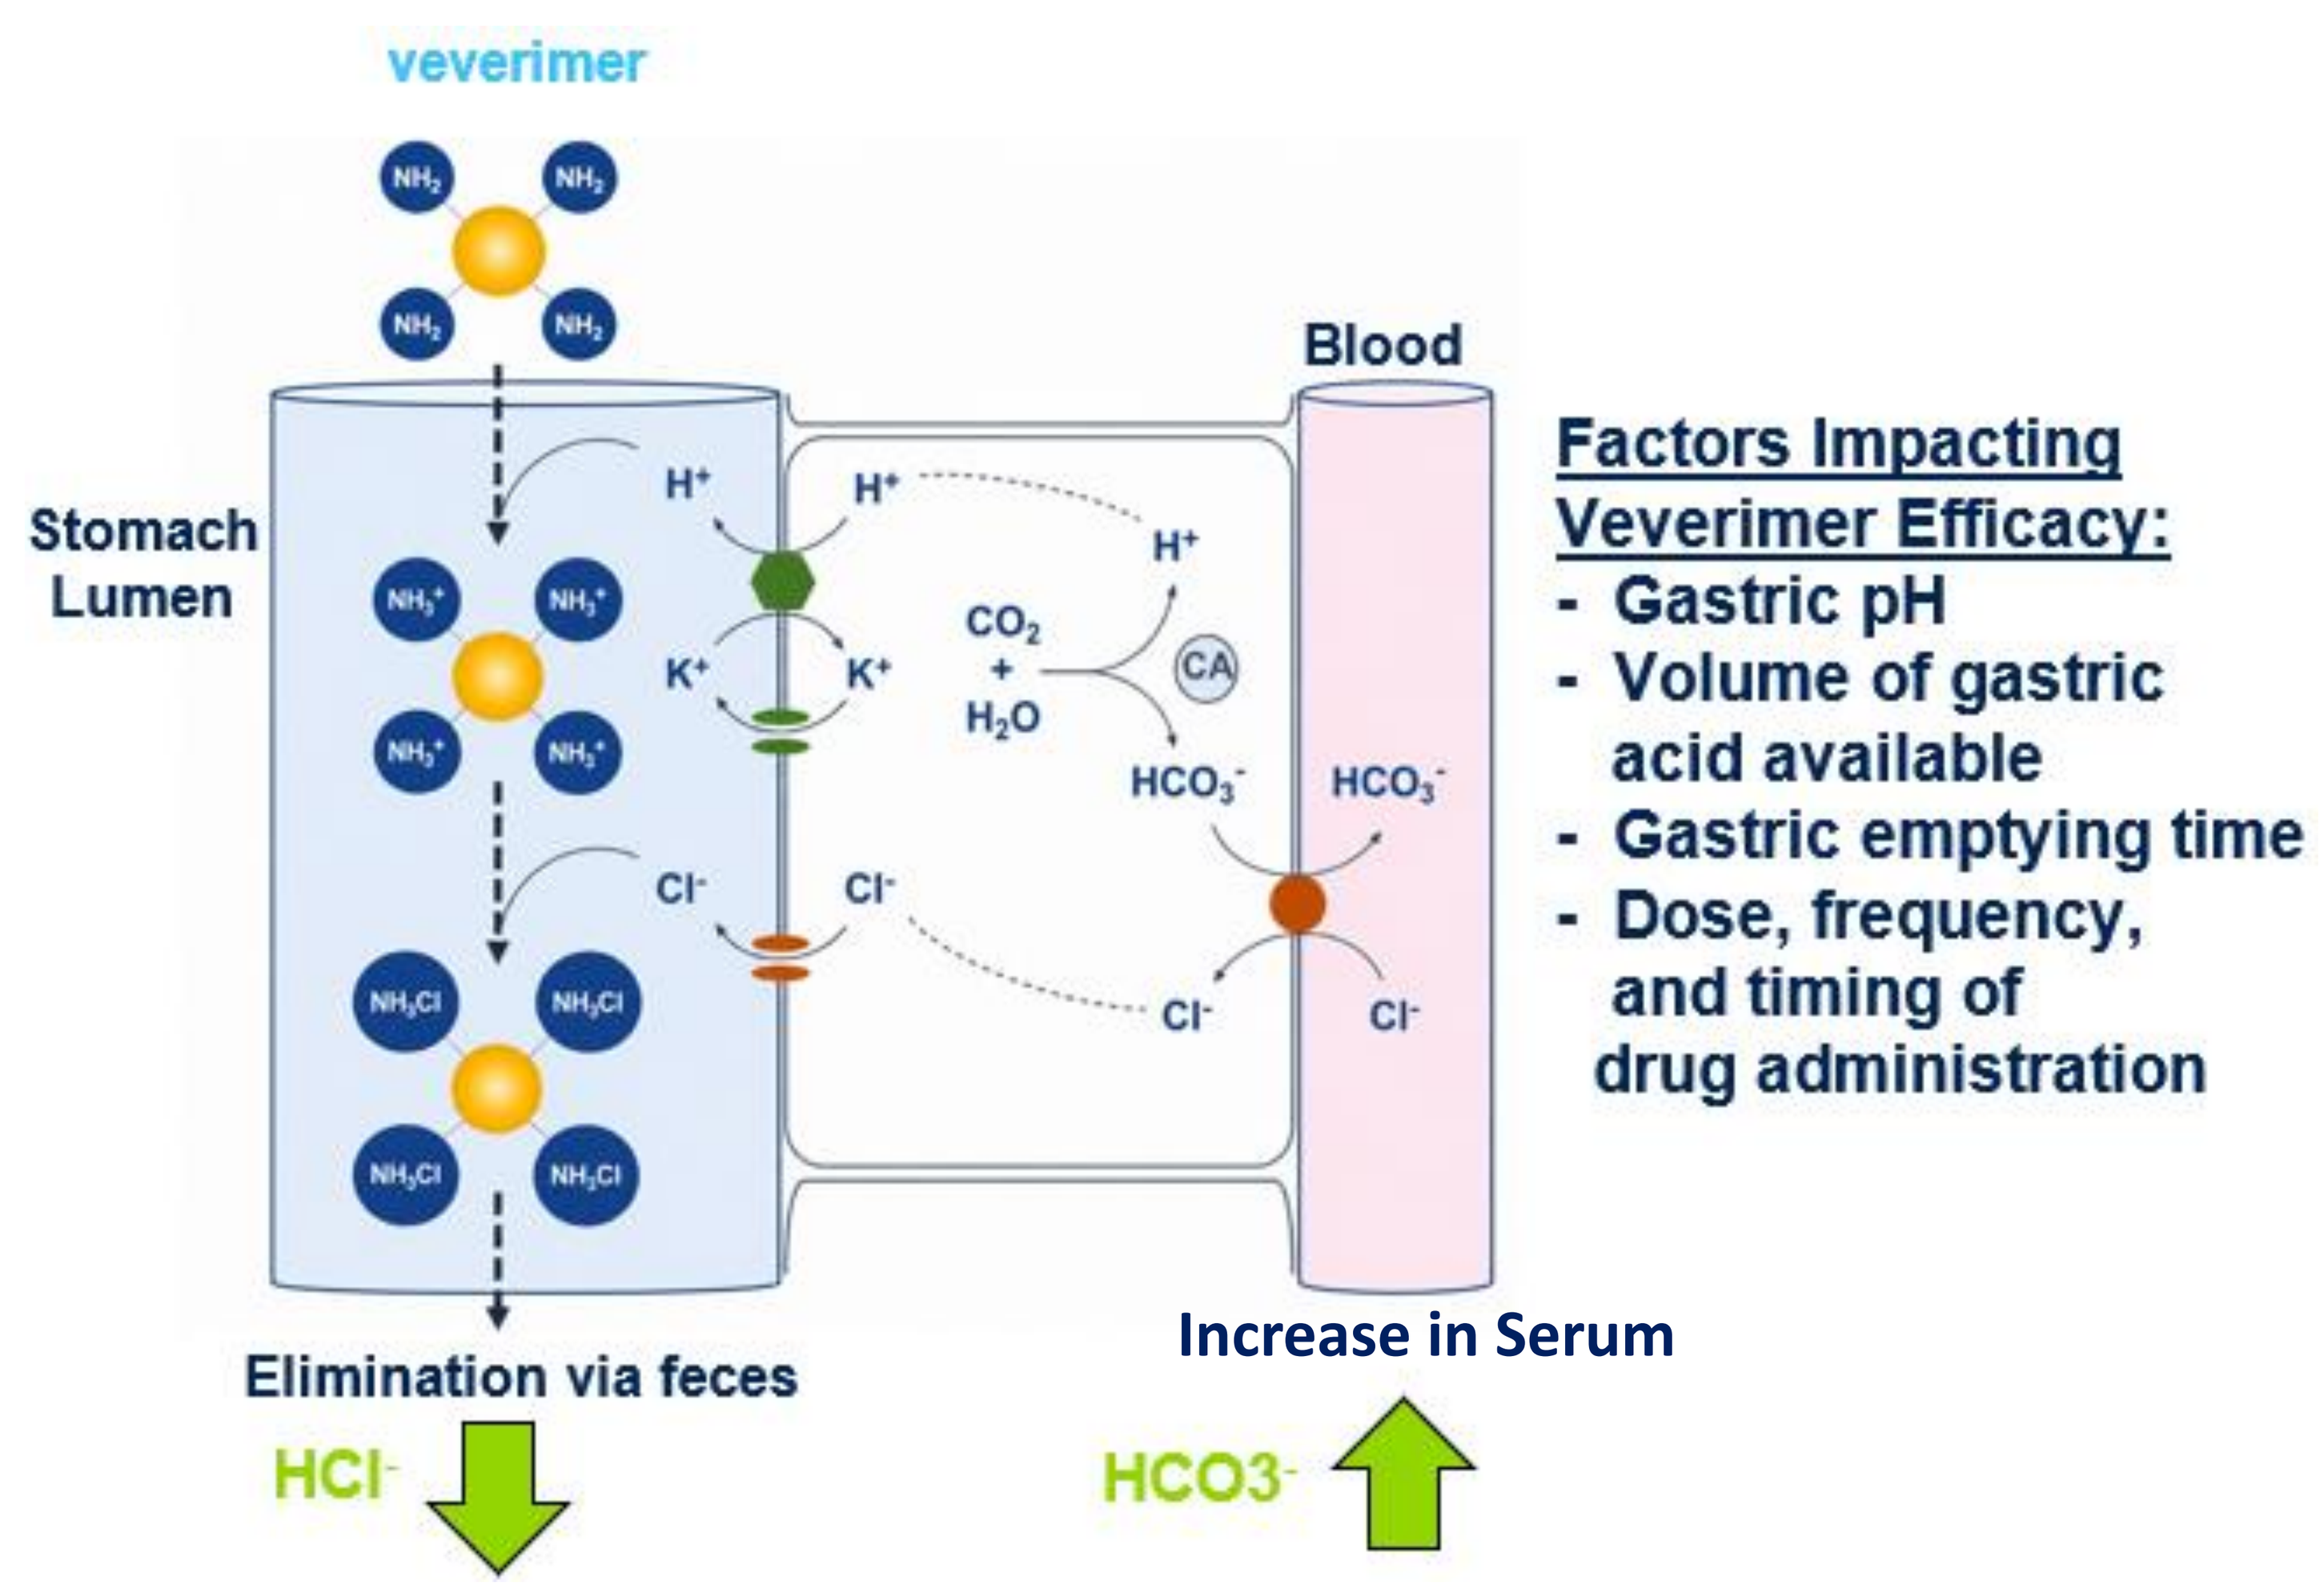
veverimer
Blood
Stomach Lumen
H^+
K^+
Cl^-
CO_2 + H_2O
CA
HCO_3^-
Factors Impacting Veverimer Efficacy:
- Gastric pH
- Volume of gastric acid available
- Gastric emptying time
- Dose, frequency, and timing of drug administration
Elimination via feces
Increase in Serum
HCl^-
$HCO3^-$

B. Increased Gastric pH by Certain Medications or Medical Conditions Will Reduce HCl Availability

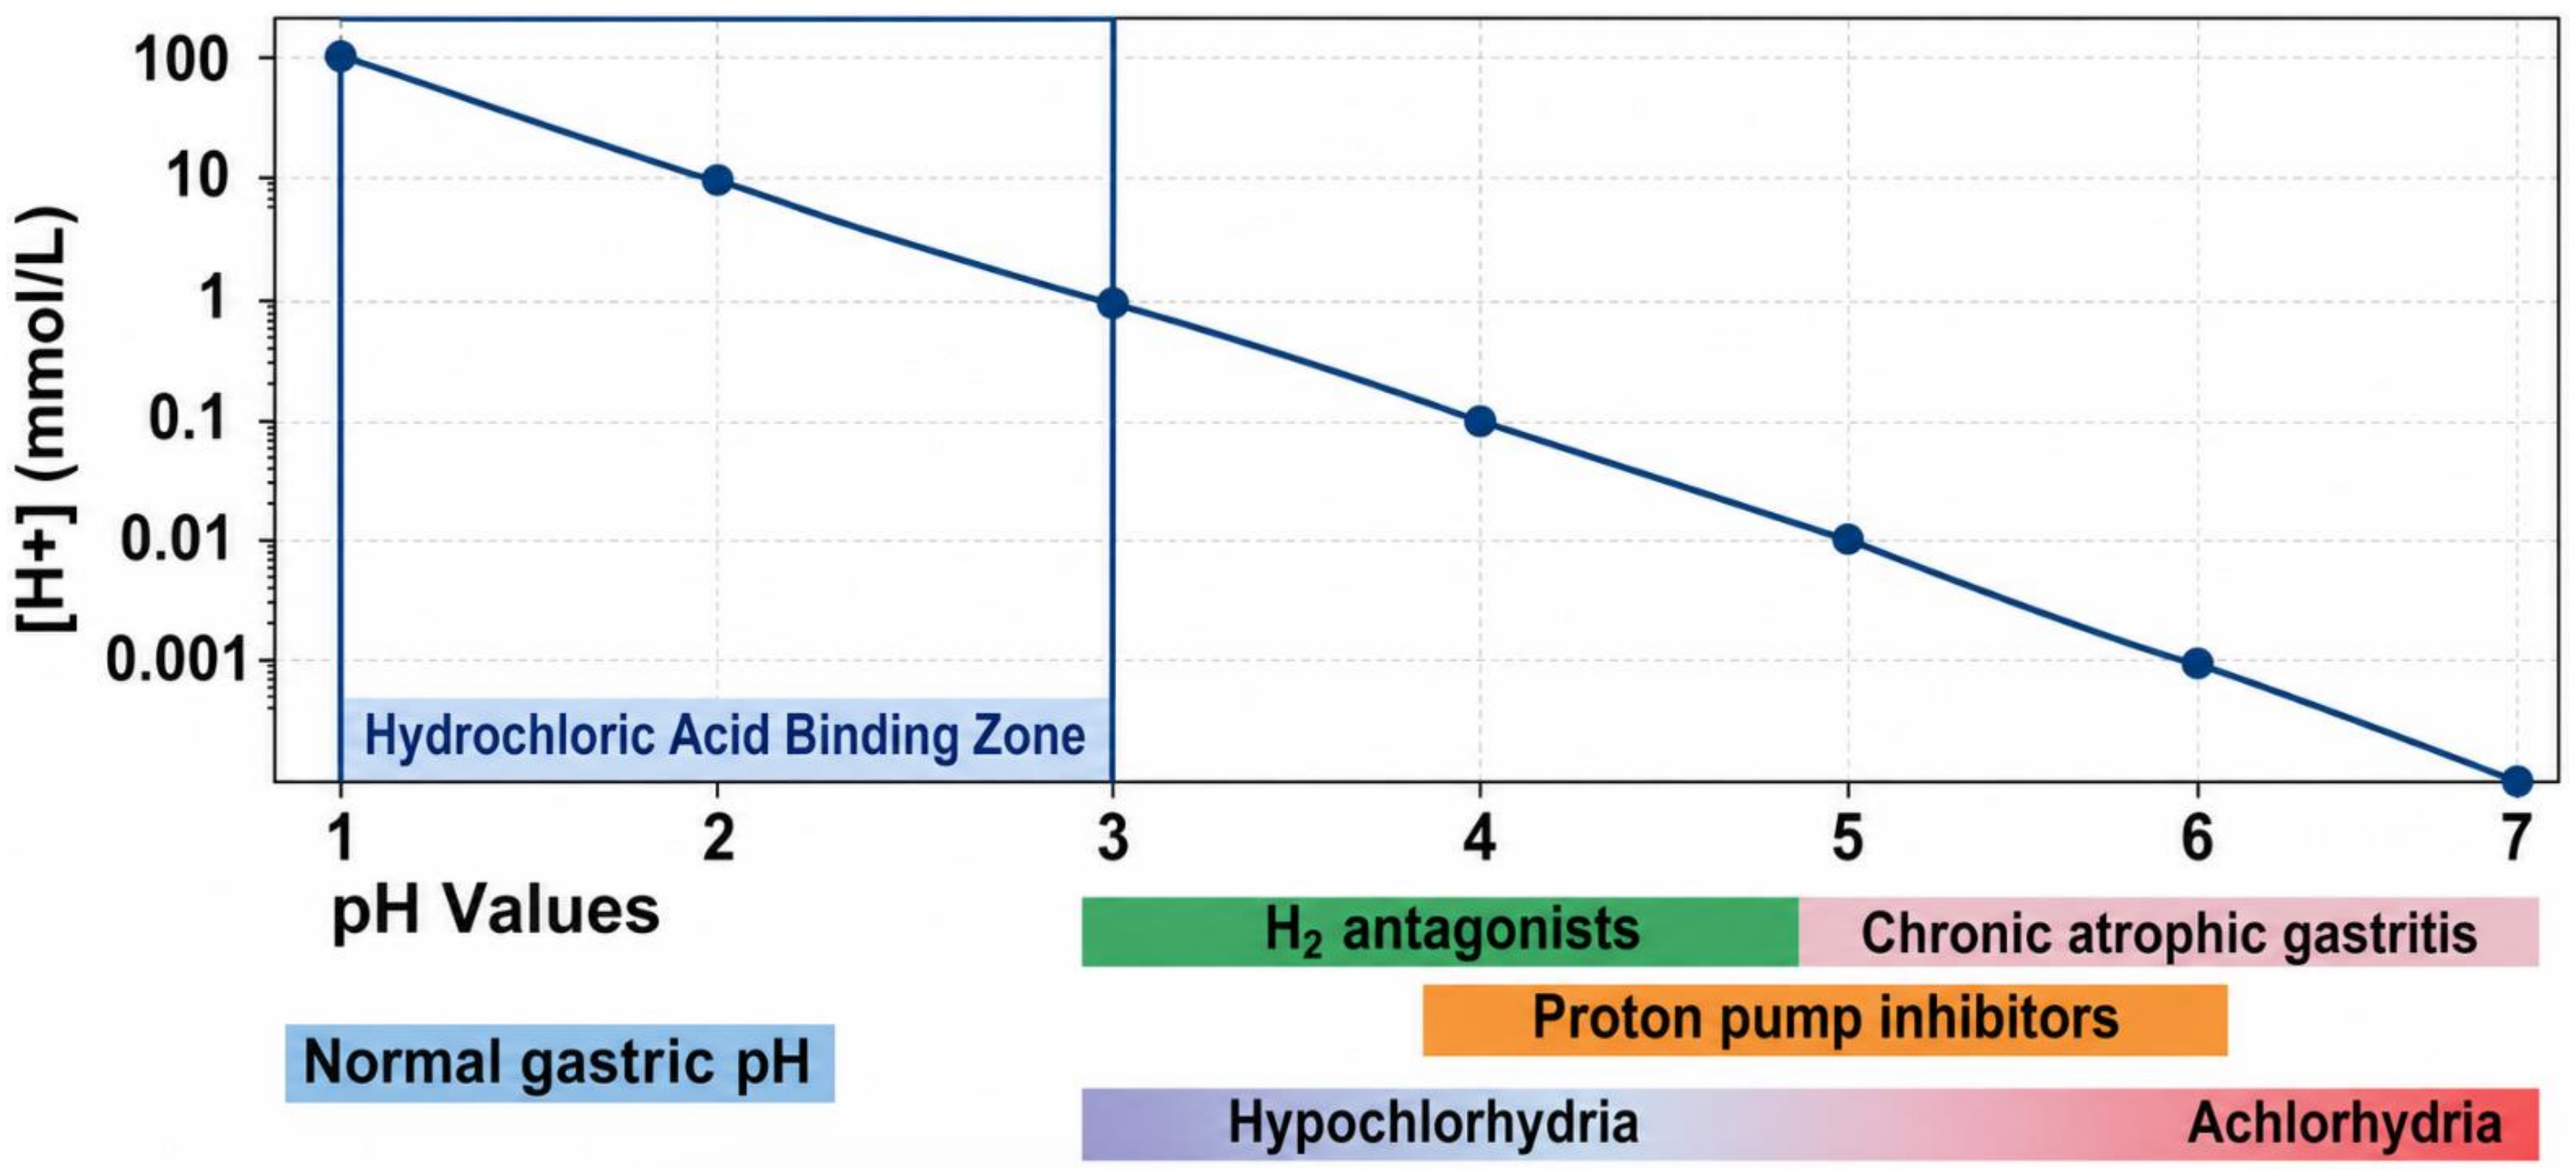
[H+] (mmol/L)
100
10
1
0.1
0.01
0.001
Hydrochloric Acid Binding Zone
1
2
3
4
5
6
7
pH Values
Normal gastric pH
H_2 antagonists
Chronic atrophic gastritis
Proton pump inhibitors
Hypochlorhydria
Achlorhydria

C. Increased Dose Frequency and Administration with Larger Meals May Improve Veverimer Efficacy

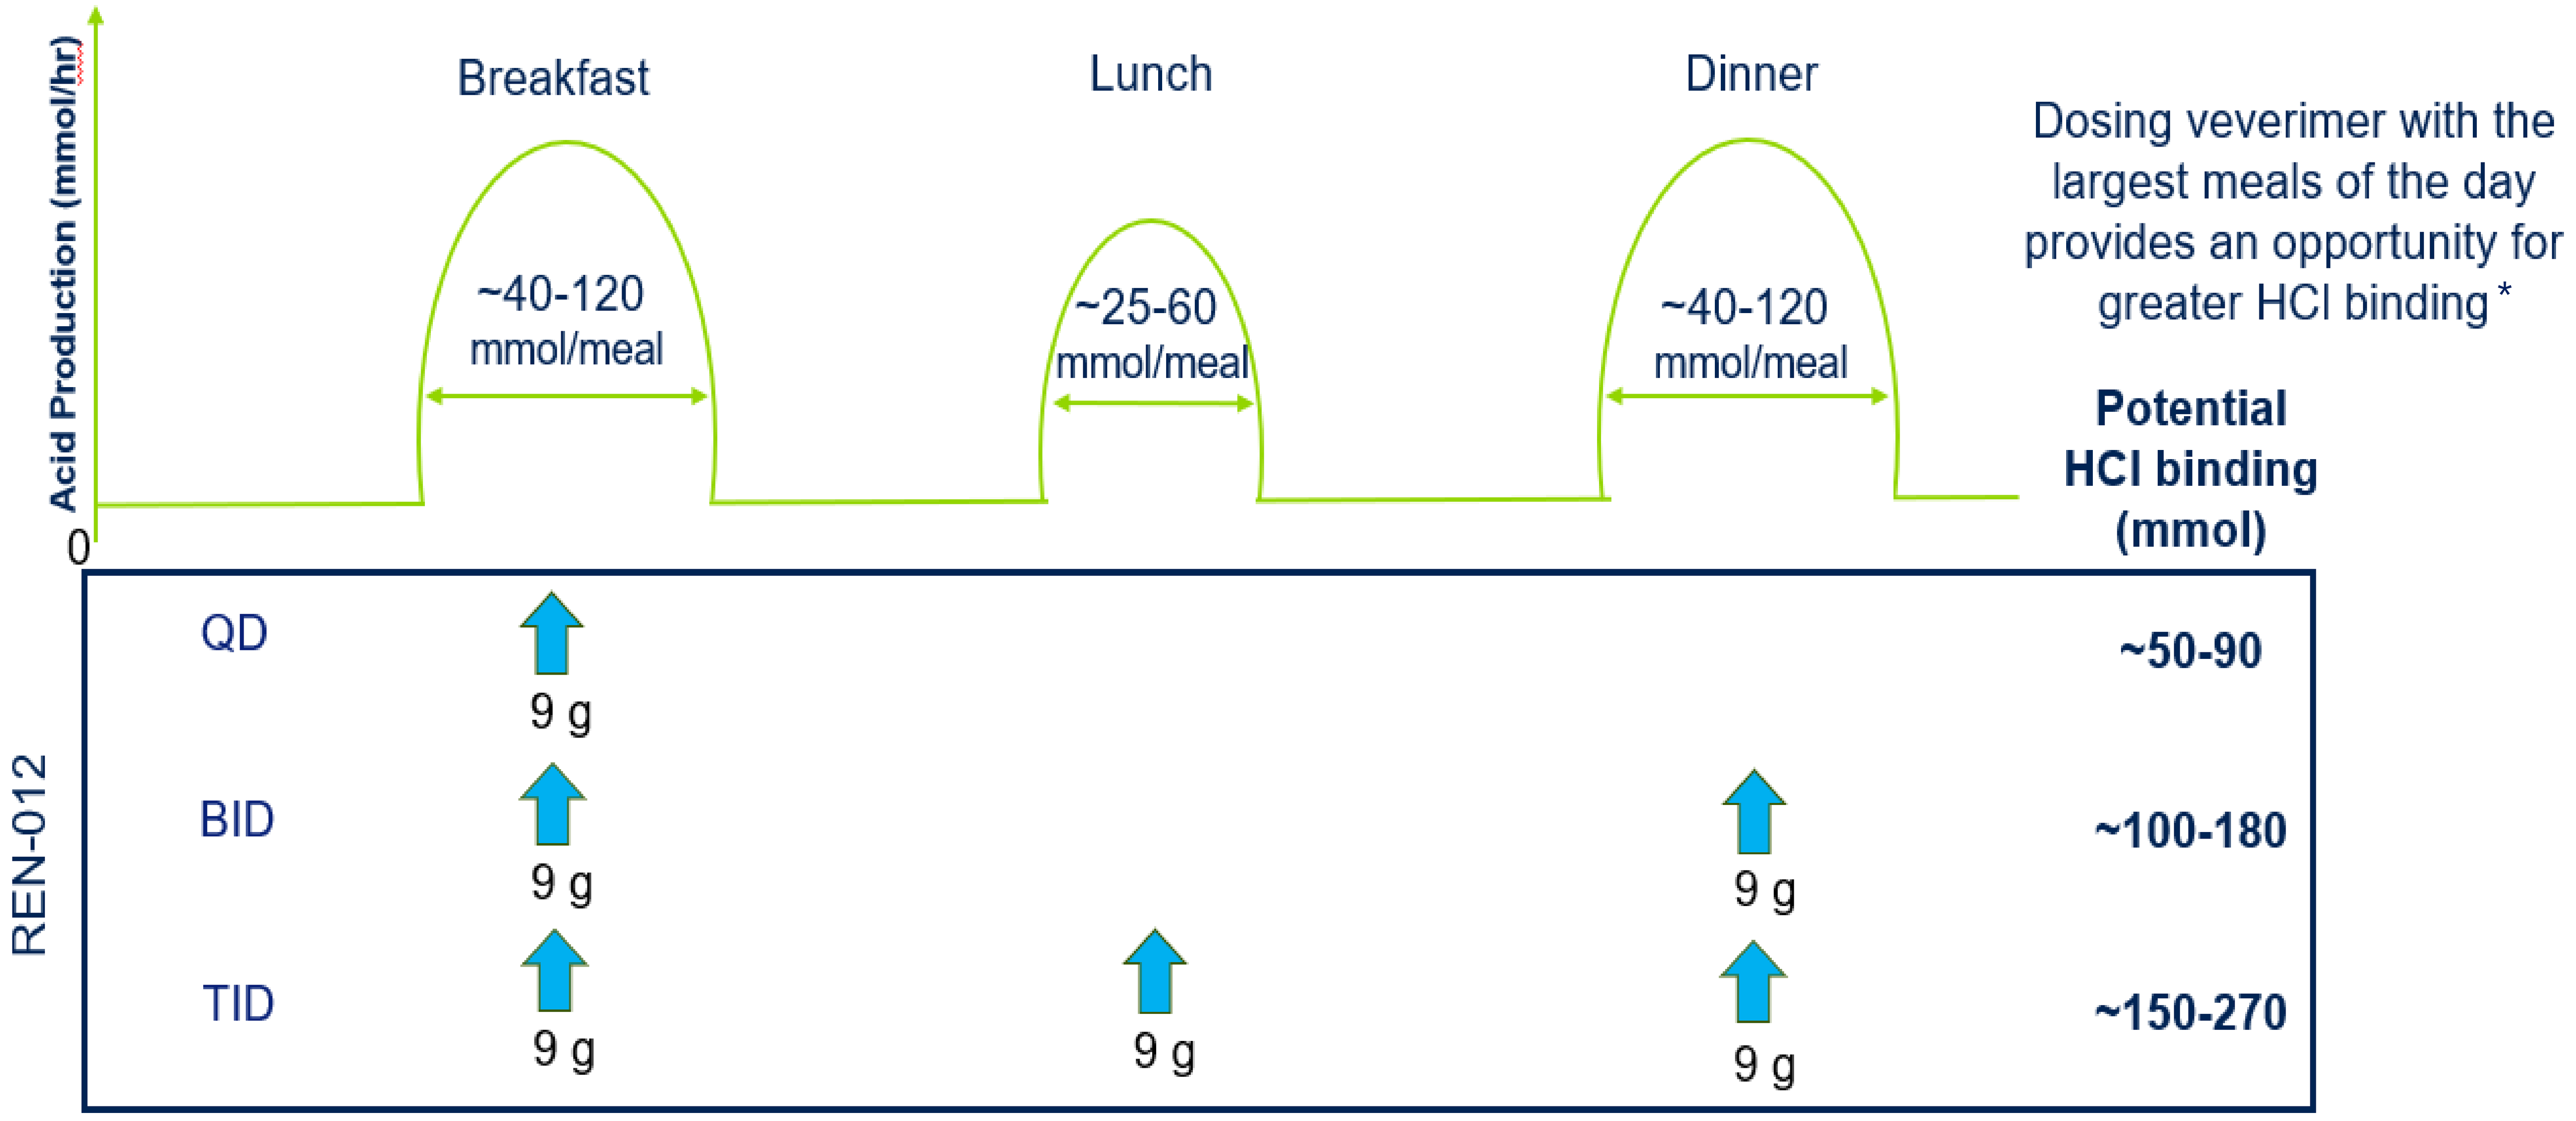
Acid Production (mmol/hr)
0
Breakfast
Lunch
Dinner
~40-120 mmol/meal
~25-60 mmol/meal
~40-120 mmol/meal
Dosing veverimer with the largest meals of the day provides an opportunity for greater HCl binding *
Potential HCl binding (mmol)
REN-012
QD
BID
TID
9 g
~50-90
~100-180
~150-270

* Protein and fat contents of large meals will stimulate gastric acid production and prolong emptying time., respectively, thus increasing HCl availability and exposure time

CONCLUSIONS

- Management of metabolic acidosis in patients with CKD can be achieved with alkali supplementation, or acid-removal (e.g., HCl binding with veverimer)
- Alignment of the therapeutic mechanism of veverimer with gastric physiology is important for successful clinical outcomes
- Translational analyses has identified gastric pH, meal content, gastric emptying time, and dosing frequency as modifiable determinants of veverimer efficacy
- A Phase 3 study of veverimer which prospectively integrates these principles into the protocol has been initiated in 2026.

ERA LEADING EUROPEAN NEPHROLOGY